Potential Role for DNA-PKcs in CD8 T cells Infiltrating Renal Allografts in T-Cell Mediated Rejection

Owen W. Stephens, BS, MB(ASCP)CM and Shana M. Coley, MD, PhD
Multiplex Imaging Center, Arkana Laboratories, Little Rock, Arkansas, USA
Disclosures: None

Arkana Laboratories

BACKGROUND

DNA-dependent protein kinase (DNA-PKcs) has a known role in T cell development, and recent work has underscored its role in mature T cell activation and effector functioning. Given the activity of DNA-PKcs in repair of DNA breaks, its expression in recently activated T cells, and the observation that recipient CD8 T cells are largely absent from skin allografts in mice lacking DNA-PKcs activity, we hypothesized that DNA-PKcs may be important for maintaining nuclear integrity of CD8 T cells during migration through peripheral tissues following activation. As a first step, we sought to determine whether or not DNA-PKcs is expressed in CD8 T cells infiltrating human kidney allografts, specifically in the setting of T-cell mediated rejection (TCMR).

METHODS

We used quantitative multiplex immunofluorescence microscopy to characterize renal cortical CD8 T cell infiltrates in whole slide scans of post-diagnostic FFPE needle core biopsies of human kidney allografts showing either TCMR (n=3) or No Rejection (n=3). Automated multiplex immunofluorescent staining for CD8, CD20, CD68, PD-1, and DNA-PKcs (for cell phenotyping) and collagen III/IV (for spatial context) was performed on a Leica BondRx following standard Akoya Biosystems MOTiF Opal workflows (see Figure 1). Image acquisition and spectral unmixing were by PhenoImager HT and InForm (version 2.6), both Akoya Biosciences. Whole biopsy images were analyzed using Halo AI and the HiPlexFL module (Indica Labs).

Target/Antigen	1° Antibody Clone	Opal/Fluor	Opal Pseudocolor
Nuclei	n/a	DAPI	
CD8	4B11	480	
PD-1	EPR4877(2)	520	
CD20	L26	570	
DNA-PKcs	polyclonal	620	
Collagen III/IV	EPR24255-13 / EPR20966	690	
CD68	PG-M1	780	

Workflow for Multiplex IF Image Analysis

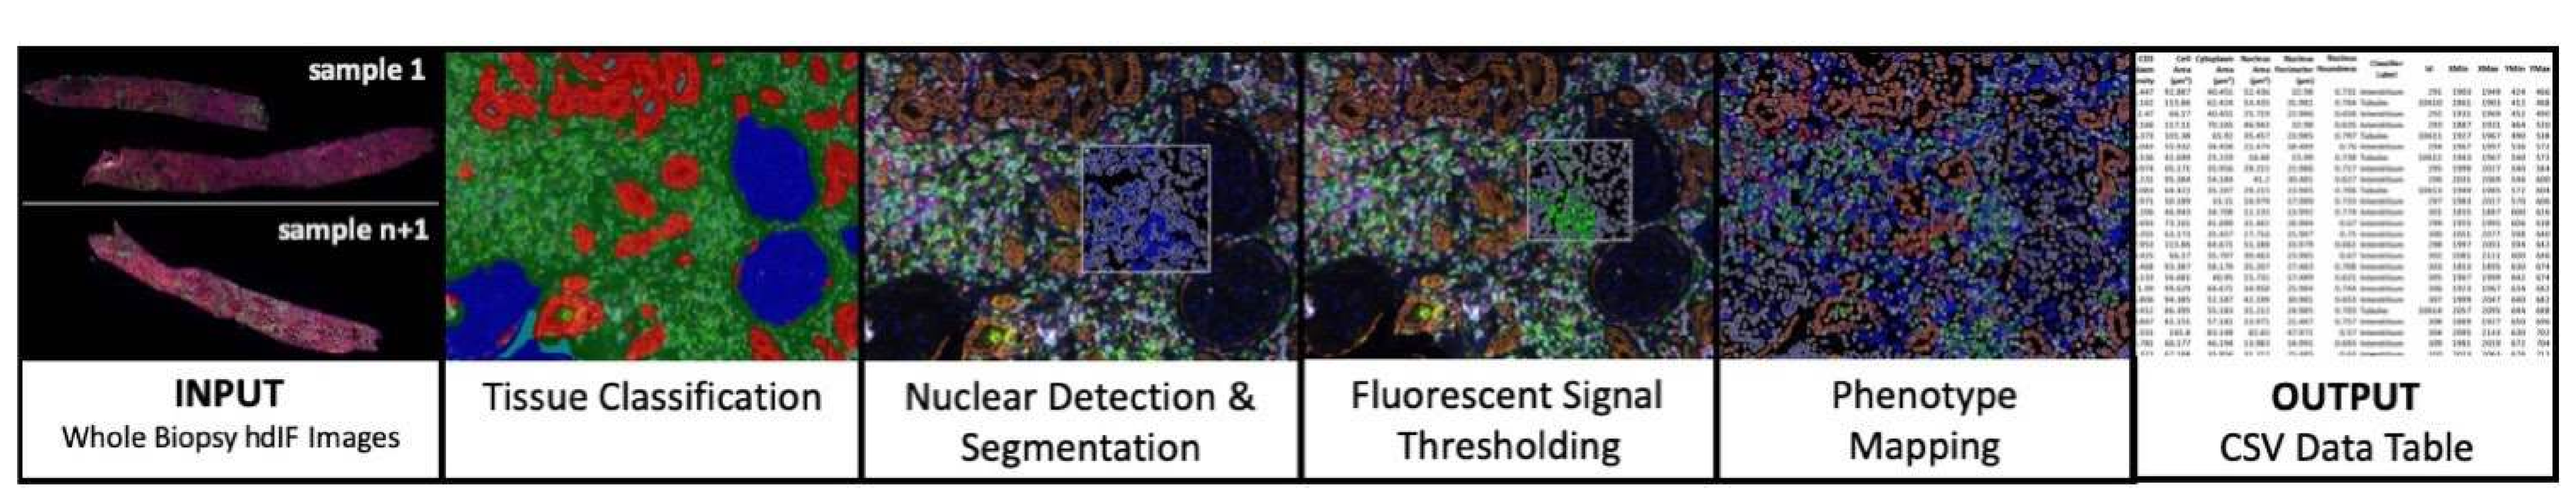
sample 1
sample n+1
INPUT
Whole Biopsy hdIF Images
Tissue Classification
Nuclear Detection & Segmentation
Fluorescent Signal Thresholding
Phenotype Mapping
OUTPUT
CSV Data Table

RESULTS

In human kidney allograft biopsies, the CD8 T cell proportion increased in TCMR to 2.64% of cells from 0.59% within No Rejection controls, excluding all other leukocytes. Along with the increase in the proportion of CD8 T cells in the kidney tissue in the setting of TCMR, there also was an increase in the proportion of DNA-PKcs positivity within the infiltrating CD8 T cell population (49.02% DNA-PKcs+ CD8 T cells in TCMR; 35.38% DNA-PKcs+ CD8 T cells in No Rejection controls).

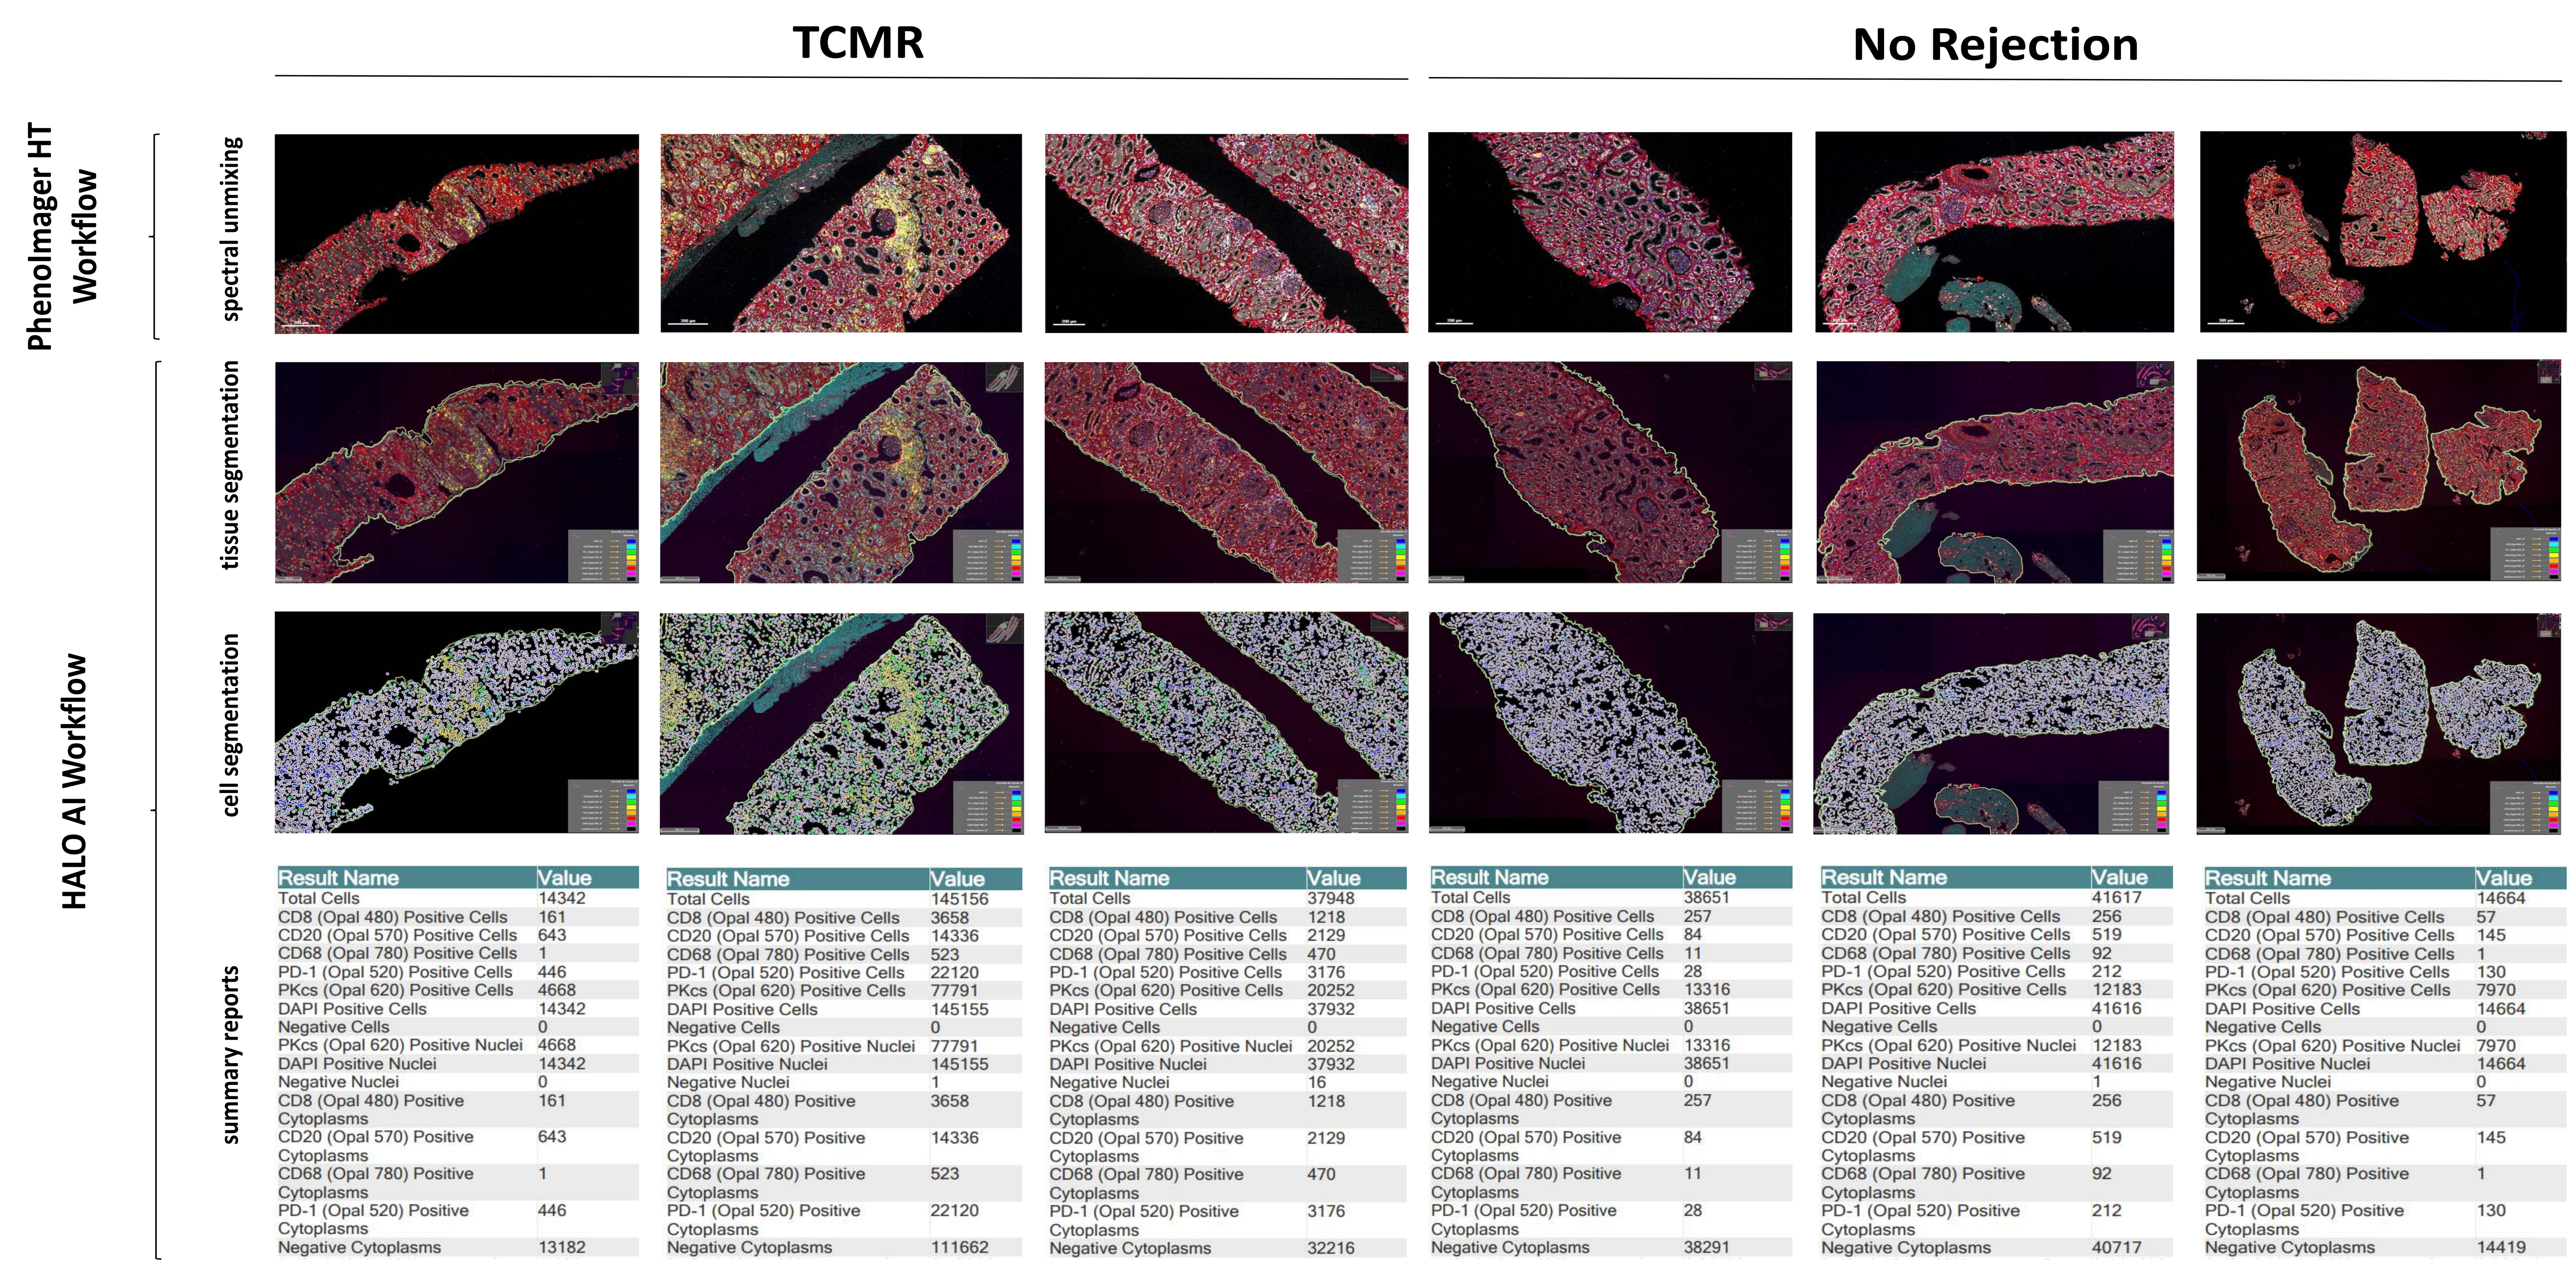
TCMR
No Rejection
PhenoImager HT Workflow
spectral unmixing
tissue segmentation
cell segmentation
HALO AI Workflow
summary reports

Result Name	Value	Value	Value	Value	Value	Value
Total Cells	14342	145156	37948	38651	41617	14664
CD8 (Opal 480) Positive Cells	161	3658	1218	257	256	57
CD20 (Opal 570) Positive Cells	643	14336	2129	84	519	145
CD68 (Opal 780) Positive Cells	1	523	470	11	92	1
PD-1 (Opal 520) Positive Cells	446	22120	3176	28	212	130
PKcs (Opal 620) Positive Cells	4668	77791	20252	13316	12183	7970
DAPI Positive Cells	14342	145155	37932	38651	41616	14664
Negative Cells	0	0	0	0	0	0
PKcs (Opal 620) Positive Nuclei	4668	77791	20252	13316	12183	7970
DAPI Positive Nuclei	14342	145155	37932	38651	41616	14664
Negative Nuclei	0	1	16	0	1	0
CD8 (Opal 480) Positive Cytoplasms	161	3658	1218	257	256	57
CD20 (Opal 570) Positive Cytoplasms	643	14336	2129	84	519	145
CD68 (Opal 780) Positive Cytoplasms	1	523	470	11	92	1
PD-1 (Opal 520) Positive Cytoplasms	446	22120	3176	28	212	130
Negative Cytoplasms	13182	111662	32216	38291	40717	14419

CONCLUSIONS

CD8 T cells infiltrating human kidney allografts express DNA-PKcs. The proportion of the intra-allograft CD8 T cell population that expresses DNA-PKcs appears related to the extent of allograft infiltration by CD8 T cells. Therefore, DNA-PKcs may warrant further investigation of its role in CD8 T cells during graft rejection and evaluation as a potential novel therapeutic target for treatment of TCMR.

REFERENCES

1. Harrison DK, Waldrip ZJ, Burdine L, Shalin SC, Burdine MS. DNA-PKcs Inhibition Extends Allogeneic Skin Graft Survival. Transplantation. 2021 Mar 1;105(3):540-549. doi: 10.1097/TP.0000000000003442. PMID: 32890138; PMCID: PMC7902289.
2. Waldrip ZJ, Burdine L, Harrison DK, Azevedo-Pouly AC, Storey AJ, Moffett OG, Mackintosh SG, Burdine MS. DNA-PKcs kinase activity stabilizes the transcription factor Egr1 in activated immune cells. J Biol Chem. 2021 Oct;297(4):101209. doi: 10.1016/j.jbc.2021.101209. Epub 2021 Sep 23. PMID: 34562454; PMCID: PMC8551498.
3. Yue X, Bai C, Xie D, Ma T, Zhou PK. DNA-PKcs: A Multi-Faceted Player in DNA Damage Response. Front Genet. 2020 Dec 23;11:607428. doi: 10.3389/fgene.2020.607428. PMID: 33424929; PMCID: PMC7786053.